9526

Phase 2 Study of Cemiplimab, a Human Monoclonal Anti–PD-1, in Patients with Metastatic Cutaneous Squamous Cell Carcinoma (Group 1): 12-Month Follow-Up

Alexander Guminski,[1] Annette M. Lim,[2] Nikhil I. Khushalani,[3] Chrysalyne D. Schmults,[4] Leonel Hernandez-Aya,[5] Badri Modi,[6] Lara A. Dunn,[7] Brett G.M. Hughes,[8] Anne Lynn S. Chang,[9] Axel Hauschild,[10] Michael R. Migden,[11] Ralf Gutzmer,[12] Murad Alam,[13] Elizabeth Stankevich,[14] Jocelyn Booth,[15] Siyu Li,[15] Israel Lowy,[15] Matthew G. Fury,[15] Danny Rischin[16]

[1]Department of Medical Oncology, Royal North Shore Hospital, St Leonards, Australia; [2]Department of Medical Oncology, Sir Charles Gairdner Hospital, Perth, Australia; [3]Department of Cutaneous Oncology, Moffitt Cancer Center, Tampa, FL, USA; [4]Department of Dermatology, Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [5]Division of Medical Oncology, Department of Medicine, Washington University School of Medicine, St Louis, MO, USA; [6]Division of Dermatology, City of Hope, Duarte, CA, USA; [7]Department of Medicine, Head and Neck Medical Oncology, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [8]Royal Brisbane & Women's Hospital and University of Queensland, Brisbane, Australia; [9]Department of Dermatology, Stanford University School of Medicine, Redwood City, CA, USA; [10]Schleswig-Holstein University Hospital, Kiel, Germany; [11]Departments of Dermatology and Head and Neck Surgery, University of Texas MD Anderson Cancer Center, Houston, TX, USA; [12]Skin Cancer Center Hannover, Department of Dermatology and Allergy, Hannover Medical School, Hannover, Germany; [13]Department of Dermatology, Feinberg School of Medicine, Northwestern University, Chicago, IL, USA; [14]Regeneron Pharmaceuticals, Inc., Basking Ridge, NJ, USA; [15]Regeneron Pharmaceuticals, Inc., Tarrytown, NY, USA; [16]Department of Medical Oncology, Peter MacCallum Cancer Centre and University of Melbourne, Melbourne, Australia

Background

- Cutaneous squamous cell carcinoma (CSCC) is one of the most common cancers worldwide and is rivalled in incidence only by basal cell carcinoma as the most common cancer in the US.[1,2]
- Until recently, there was no approved systemic therapy for patients with advanced CSCC, a term that comprises metastatic and locally advanced CSCC not amenable to surgery and/or radiotherapy.
- Cemiplimab is a high affinity, human, hinge-stabilized IgG4 monoclonal antibody to the programmed cell death (PD)-1 receptor that potently blocks the interactions of PD-1 with PD-ligand 1 (PD-L1) and PD-ligand 2 (PD-L2).[3]
 - In the US, cemiplimab-rwlc is the only Food and Drug Administration-approved treatment for patients with metastatic CSCC or locally advanced CSCC who are not candidates for curative surgery or curative radiation.[4]
- Primary analysis (October 2017) of cemiplimab in patients with metastatic CSCC (Group 1) in a Phase 2 study (EMPOWER-CSCC-1; NCT02760498) demonstrated substantial antitumor activity, durable responses, and acceptable safety profile.[5]
- We now report 12-month follow-up data from this group of patients.

Objectives

- The primary objective of the Phase 2 study was to evaluate objective response rate (ORR; complete response + partial response according to independent central review [ICR]) per Response Evaluation Criteria In Solid Tumors (RECIST) 1.1[6] (for scans) and modified World Health Organization (WHO) criteria (for photos).
- Secondary objectives included estimation of ORR by investigator assessments (INV), duration of response, progression-free survival (PFS), overall survival (OS), and assessment of safety and tolerability of cemiplimab.
 - Durable disease control rate (defined as the proportion of patients without progressive disease for at least 105 days) was also assessed.

Methods

- Adult patients with metastatic CSCC (nodal and/or distant) from Group 1 of EMPOWER-CSCC-1, a Phase 2, non-randomized, global, pivotal trial of cemiplimab in patients with advanced CSCC are included in this analysis (**Figure 1**).
- Severity of treatment-emergent adverse events (TEAEs) was graded according to the National Cancer Institute Common Terminology Criteria for Adverse Events (version 4.03).
- The data cut-off date for this analysis was September 20, 2018.

Figure 1. EMPOWER-CSCC-1 study design (NCT02760498)

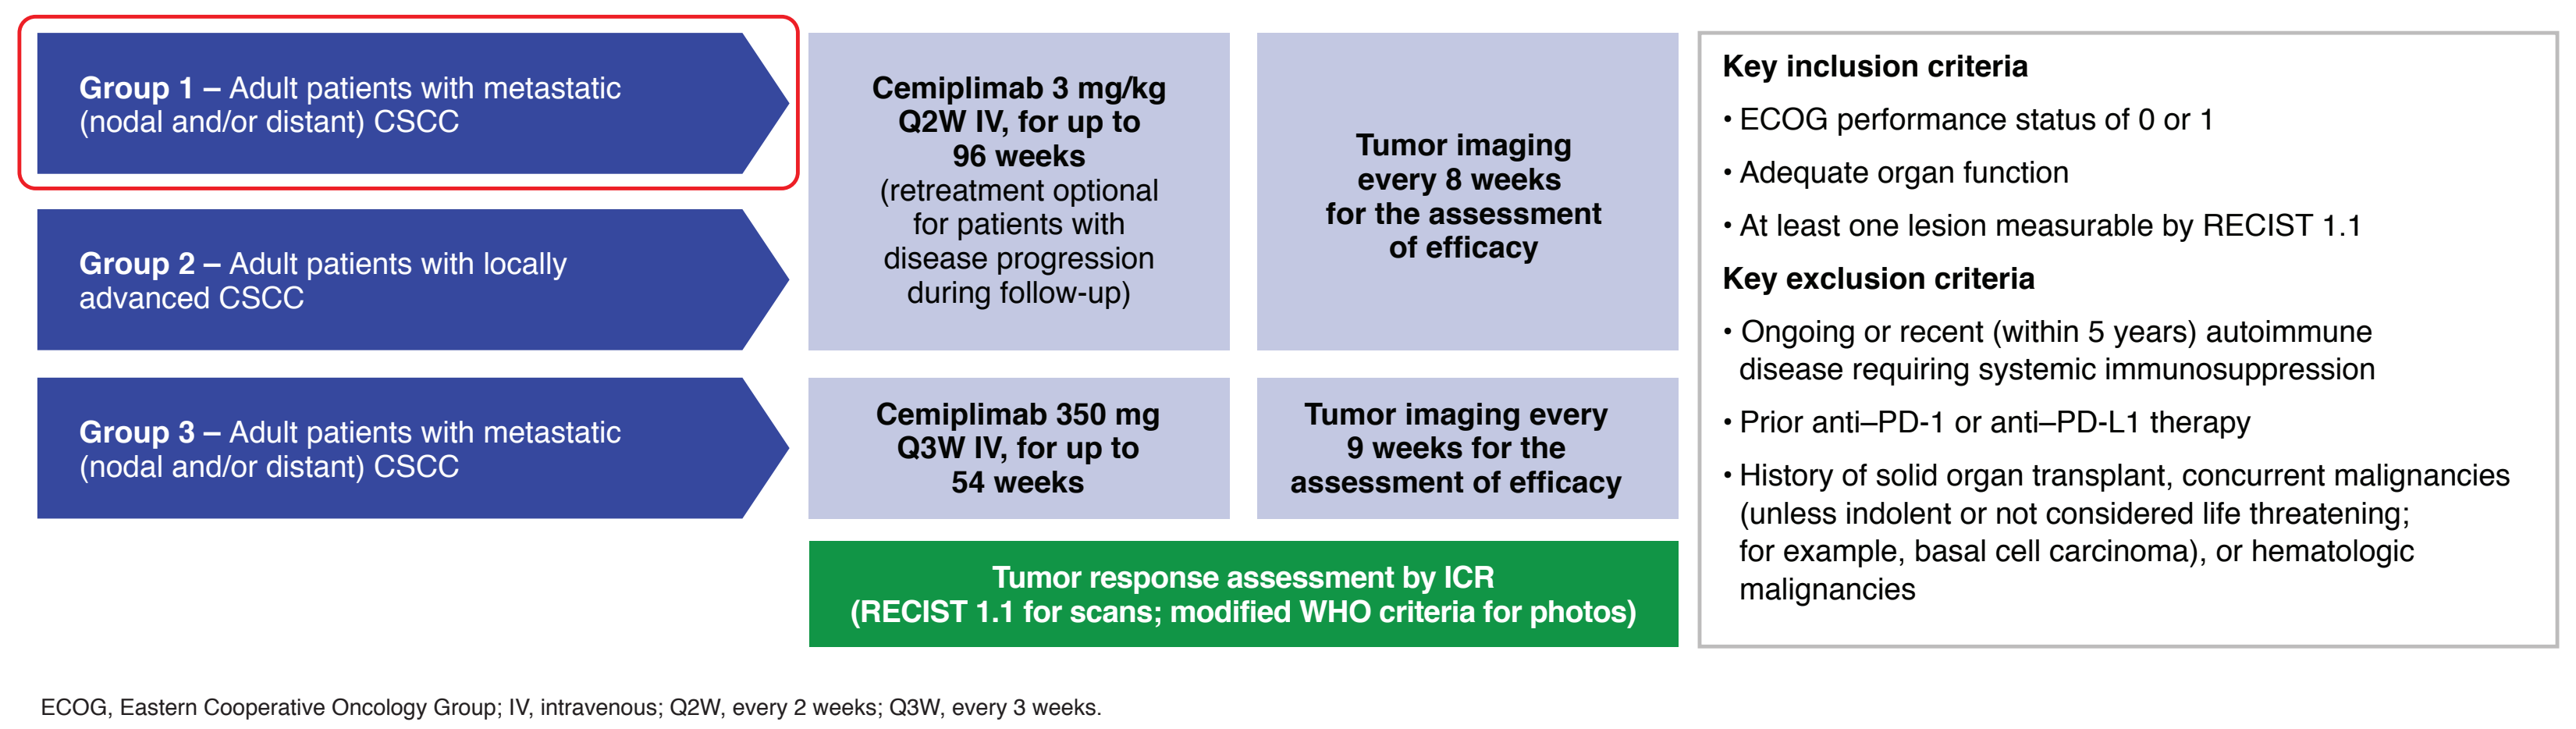
Group 1 – Adult patients with metastatic (nodal and/or distant) CSCC
Group 2 – Adult patients with locally advanced CSCC
Group 3 – Adult patients with metastatic (nodal and/or distant) CSCC
Cemiplimab 3 mg/kg Q2W IV, for up to 96 weeks (retreatment optional for patients with disease progression during follow-up)
Tumor imaging every 8 weeks for the assessment of efficacy
Cemiplimab 350 mg Q3W IV, for up to 54 weeks
Tumor imaging every 9 weeks for the assessment of efficacy
Tumor response assessment by ICR (RECIST 1.1 for scans; modified WHO criteria for photos)
Key inclusion criteria
• ECOG performance status of 0 or 1
• Adequate organ function
• At least one lesion measurable by RECIST 1.1
Key exclusion criteria
• Ongoing or recent (within 5 years) autoimmune disease requiring systemic immunosuppression
• Prior anti–PD-1 or anti–PD-L1 therapy
• History of solid organ transplant, concurrent malignancies (unless indolent or not considered life threatening; for example, basal cell carcinoma), or hematologic malignancies

ECOG, Eastern Cooperative Oncology Group; IV, intravenous; Q2W, every 2 weeks; Q3W, every 3 weeks.

Results

Baseline characteristics, disposition, and treatment exposure

- A total of 59 patients were enrolled and treated with cemiplimab 3 mg/kg Q2W (**Table 1**).
- At the time of data cut-off, 13 patients (22.0%) had completed the planned treatment, 13 (22.0%) remained on treatment, and 33 (55.9%) had discontinued treatment mainly due to disease progression (n=19; 32.2%) and adverse events (n=6; 10.2%). Two of the remaining eight patients who had discontinued treatment had done so due to complete response to cemiplimab.
- The median duration of exposure to cemiplimab was 14.9 months (range: 0.5–22.1) and the median number of doses administered was 31 (range: 1–48).
- The median duration of follow-up at the time of data cut-off was 16.5 months (range: 1.1–26.6).

Table 1. Patient demographics and baseline characteristics

	Metastatic CSCC (N=59)
Median age, years (range)	71 (38–93)
≥65 years, n (%)	43 (72.9)
Male, n (%)	54 (91.5)
ECOG performance status, n (%)	
0	23 (39.0)
1	36 (61.0)
Primary CSCC site, n (%)	
Head/neck	38 (64.4)
Extremity	12 (20.3)
Trunk	9 (15.3)
Metastasis status	
Distant	45 (76.3)
Nodal only	14 (23.7)
M stage at screening	
M0	14 (23.7)
M1	45 (76.3)
N stage at screening	
NX	9 (15.3)
N0	10 (16.9)
N1	15 (25.4)
Other†	25 (42.4)
Prior cancer-related systemic therapy, n (%)‡	33 (55.9)
Prior cancer-related radiotherapy, n (%)	50 (84.7)

†Includes N2 (n=6; 10.2%), N2B (n=4; 6.8%), N2C (n=7; 11.9%), and N3 (n=8; 13.6%). ‡Twenty-two patients had received one prior cancer-related systemic therapy and 11 had received ≥2 prior cancer-related systemic therapies.

Clinical activity

- By ICR, ORR was 49.2% (95% confidence interval [CI]: 35.9–62.5) with 10 patients experiencing a complete response and 19 experiencing a partial response (**Table 2**). By INV, ORR was also 49.2% (95% CI: 35.9–62.5; four complete responses and 25 partial responses).
- Rapid, deep, and durable reductions in target lesions were frequently observed (**Figures 2** and **3**).
- By ICR, median duration of response had not been reached at data cut-off.
 - Responses have lasted ≥12 months for 22 patients (Kaplan-Meier estimated event-free probability at 12 months in patients with confirmed complete or partial response was 88.9% [95% CI: 69.3–96.3]).
 - The longest duration of response at data cut-off was 21.6 months and was ongoing.

Table 2. Tumor response assessment by ICR

	Metastatic CSCC (N=59)
Best overall response, n (%)	
Complete response	10 (16.9)
Partial response	19 (32.2)
Stable disease	9 (15.3)
Non-complete response/non-progressive disease†	4 (6.8)
Progressive disease	10 (16.9)
Not evaluable‡	7 (11.9)
ORR, % (95% CI)§	49.2 (35.9–62.5)
Disease control rate, % (95% CI)	71.2 (57.9–82.2)
Durable disease control rate, % (95% CI)¶	62.7 (49.1–75.0)
Median observed time to response, months (range)#	1.9 (1.7–9.1)

†Patients with non-measurable disease on central review of baseline imaging. ‡Includes missing and unknown tumor response. §By INV, the ORR was 49.2% (95% CI 35.9–62.5; four complete responses and 25 partial responses). ¶Defined as the proportion of patients without progressive disease for at least 105 days. #Data shown are from patients with confirmed complete or partial response.

Figure 2. Clinical activity of tumor response to cemiplimab in patients who underwent radiologic evaluation per ICR

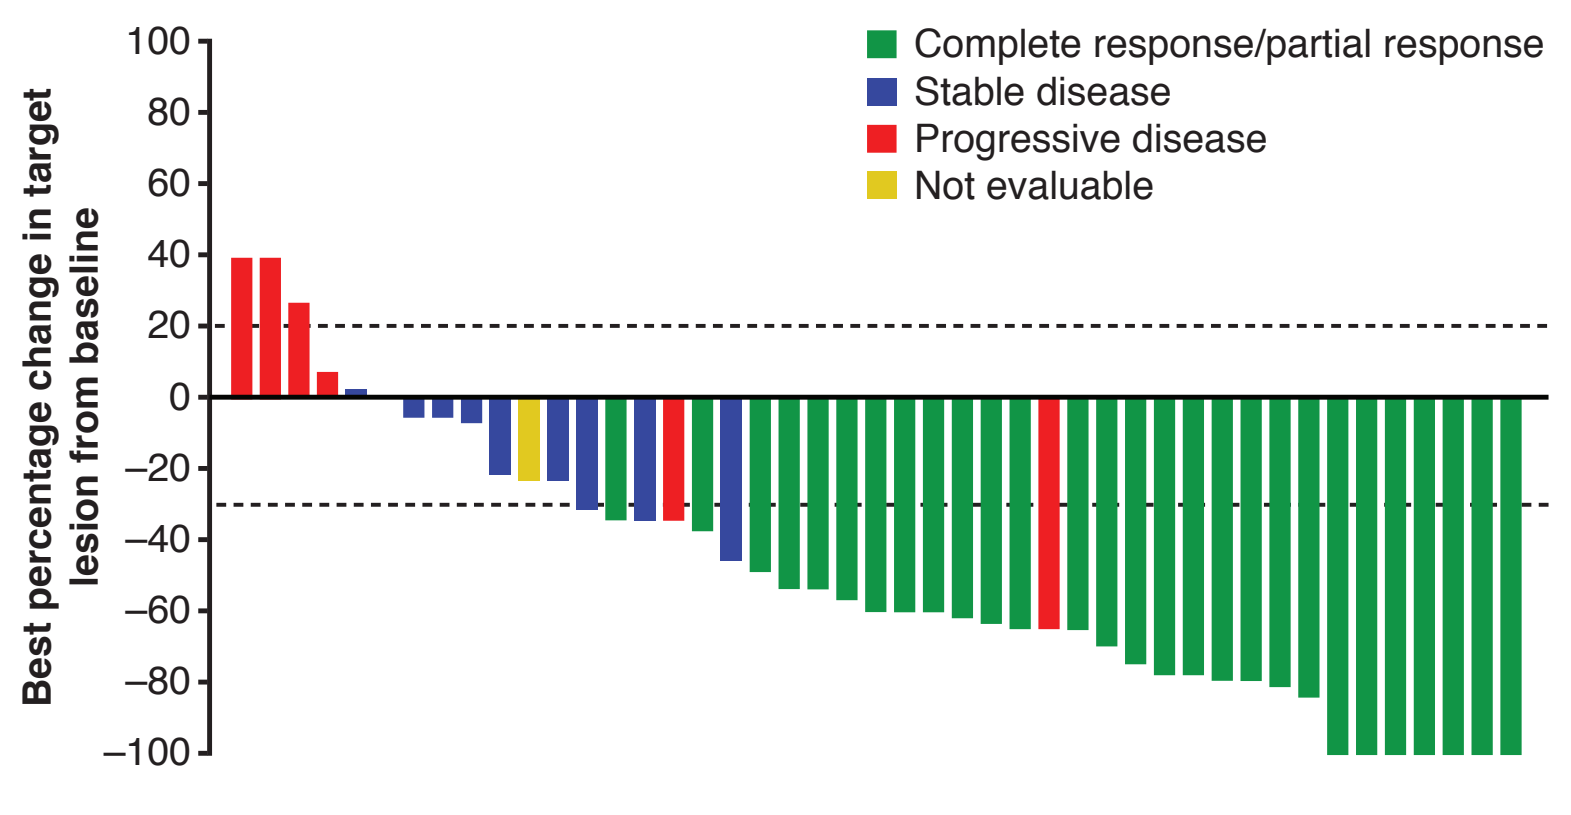
Complete response/partial response
Stable disease
Progressive disease
Not evaluable
Best percentage change in target lesion from baseline
100
80
60
40
20
0
–20
–40
–60
–80
–100

Plot shows the best percentage change in the sum of target lesion diameters from baseline for 45 patients who underwent radiologic evaluation per ICR after treatment initiation. Lesion measurements after progression were excluded. Horizontal dashed lines indicate criteria for partial response (≥30% decrease in the sum of target lesion diameters) and progressive disease (≥20% increase in the target lesion diameters). Two patients with target lesion reductions ≥30% were classified as progressive disease (red bars) due to new lesion or progression of non-target lesion. Fourteen patients do not appear in the figure (but are included in the ORR analysis [Table 2], per intention-to-treat) as they did not have baseline target lesion or evaluable post-baseline assessment. One patient had stable disease per RECIST 1.1 but was not evaluable (yellow bar) due to externally visible disease that was not evaluable by photographic assessments.

Figure 3. Time to and duration of response in responding patients

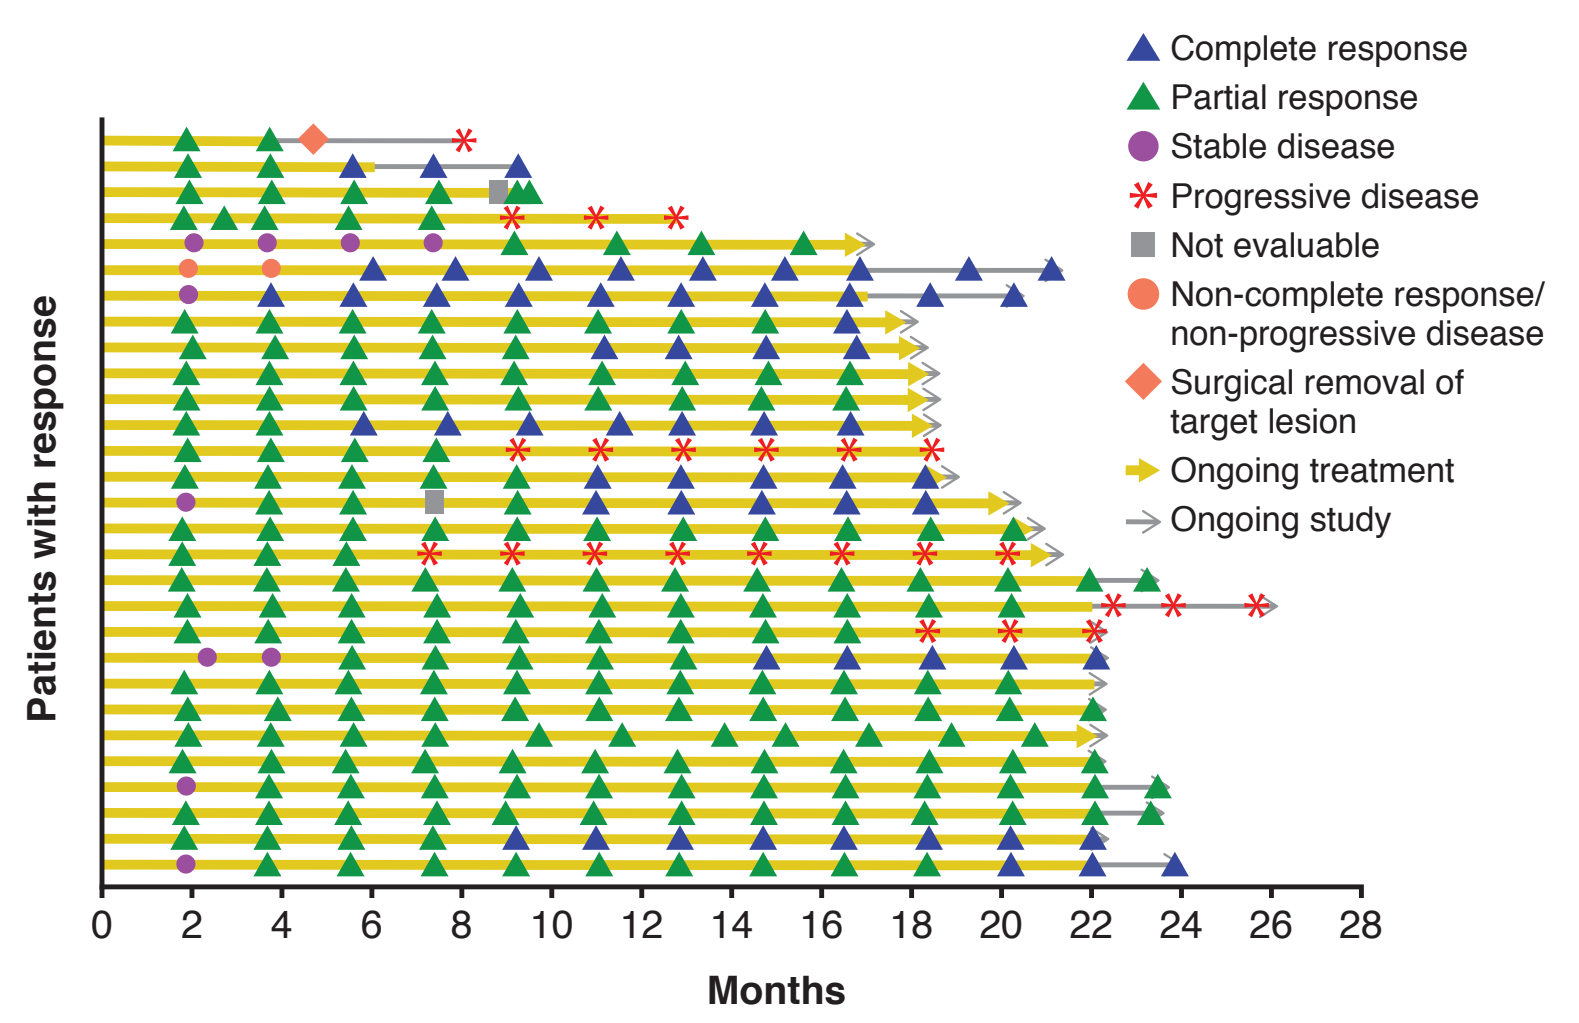
Complete response
Partial response
Stable disease
Progressive disease
Not evaluable
Non-complete response/ non-progressive disease
Surgical removal of target lesion
Ongoing treatment
Ongoing study
Patients with response
0
2
4
6
8
10
12
14
16
18
20
22
24
26
28
Months

Each horizontal line represents one patient. Twenty-three of the 29 patients remain in response at time of data cut-off; of the 23 patients, 10 were still on study, 11 were in post-treatment follow-up and two were off study. Multiple progression events for a single patient were possible due to discrepancies between INV and ICR tumor assessments and because the protocol allowed option for treatment past progression in patients whom the investigator felt were experiencing clinical benefits.

- Median PFS by ICR was 18.4 months (95% CI: 7.3–not evaluable; **Figure 4**).
- Median OS has not been reached; Kaplan-Meier estimation of OS at 24 months was 70.6% (95% CI: 57.0–80.6; **Figure 5**).

Figure 4. Kaplan–Meier curves for PFS per ICR

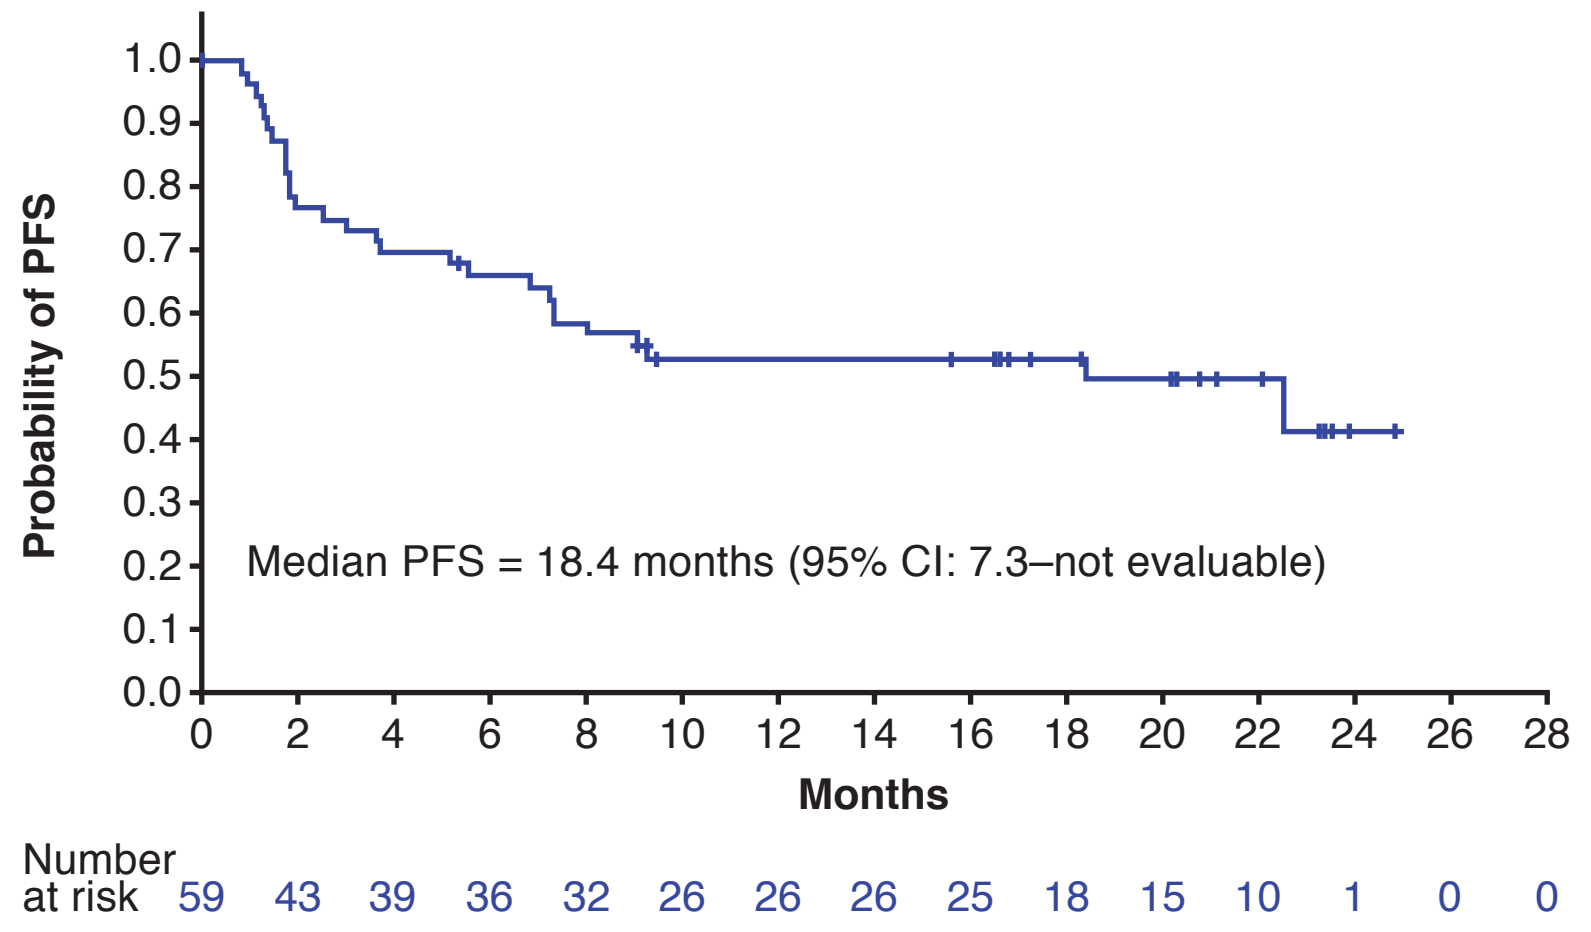
Probability of PFS
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
Median PFS = 18.4 months (95% CI: 7.3–not evaluable)
0
2
4
6
8
10
12
14
16
18
20
22
24
26
28
Months
Number at risk 59 43 39 36 32 26 26 26 25 18 15 10 1 0 0

Figure 5. Kaplan-Meier curves for OS

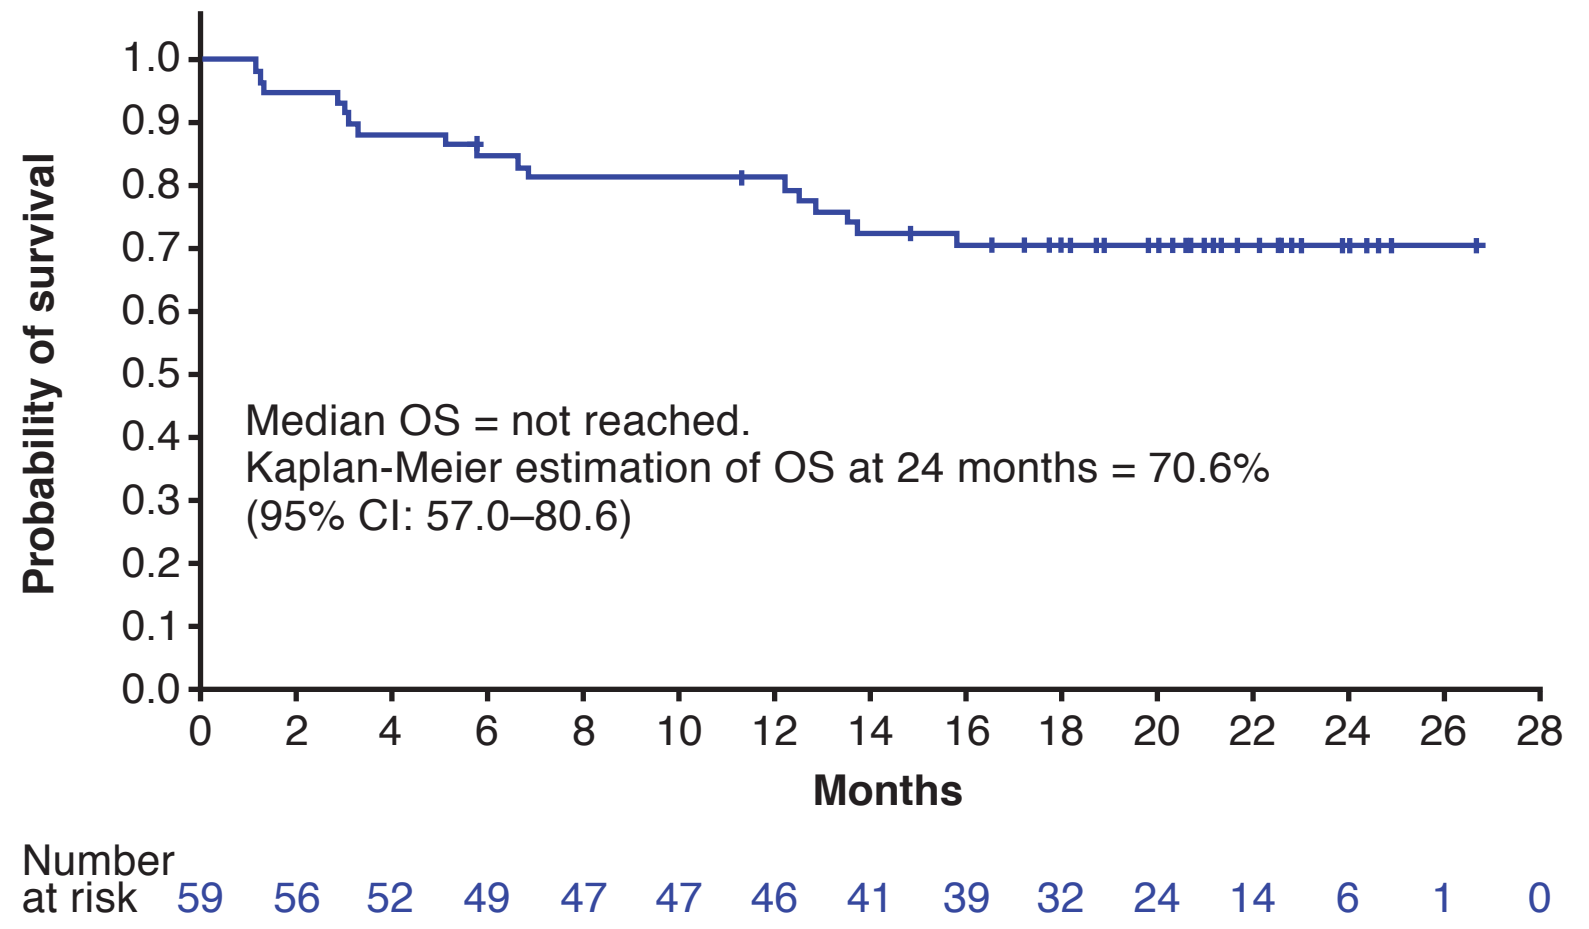
Probability of survival
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
Median OS = not reached.
Kaplan-Meier estimation of OS at 24 months = 70.6%
(95% CI: 57.0–80.6)
0
2
4
6
8
10
12
14
16
18
20
22
24
26
28
Months
Number at risk 59 56 52 49 47 47 46 41 39 32 24 14 6 1 0

Treatment-emergent adverse events

- TEAEs regardless of attribution are summarized in **Table 3**.
- Grade ≥3 TEAEs that occurred in more than one patient were cellulitis (n=4; 6.8%), pneumonitis (n=3; 5.1%), and anemia, dyspnea, hypercalcemia, new primary CSCC, pleural effusion, and pneumonia (each n=2; 3.4%).
- Grade ≥3 TEAEs that led to treatment discontinuation were pneumonitis (n=3; 5.1%) and aseptic meningitis, confusional state, and neck pain (all in the same patient: n=1; 1.7%).
- Treatment-related adverse events (TRAEs) occurred in 46 patients (78.0%) with nine patients (15.3%) experiencing the following grade ≥3 TRAEs:
 - Pneumonitis (n=3; 5.1%) and aseptic meningitis, colitis, confusional state, decreased lymphocyte count, diarrhea, duodenal ulcer, esophagitis, hypophysitis, neck pain, polyarthritis, and small intestinal hemorrhage (each n=1; 1.7%).
- Six patients (10.2%) experienced serious grade ≥3 TRAEs as follows: pneumonitis (n=3; 5.1%), and aseptic meningitis, duodenal ulcer, hypophysitis, esophagitis, and small intestine hemorrhage (each n=1; 1.7%).

Table 3. TEAEs regardless of attribution

TEAEs	Metastatic CSCC (N=59)	
n (%)	Any grade	Grade ≥3
Any	59 (100.0)	30 (50.8)
Serious	24 (40.7)	20 (33.9)
Led to discontinuation	6 (10.2)	4 (6.8)
Occurred in at least 10% of the patient population by any grade†		
Diarrhea	17 (28.8)	1 (1.7)
Fatigue	15 (25.4)	1 (1.7)
Nausea	14 (23.7)	0
Headache	11 (18.6)	0
Constipation	10 (16.9)	1 (1.7)
Pruritus	10 (16.9)	0
Rash	10 (16.9)	0
Arthralgia	9 (15.3)	0
Cough	9 (15.3)	0
Decreased appetite	8 (13.6)	0
Maculopapular rash	8 (13.6)	0
Anemia	7 (11.9)	2 (3.4)
Dizziness	7 (11.9)	0
Dry skin	6 (10.2)	0
Dyspnea	6 (10.2)	2 (3.4)
Hypothyroidism	6 (10.2)	0
Oropharyngeal pain	6 (10.2)	0
Pneumonitis	6 (10.2)	3 (5.1)
Upper respiratory tract infection	6 (10.2)	0
Vomiting	6 (10.2)	0

†Events are listed as indicated on the case report form. Although rash and maculopapular rash may reflect the same condition, they were listed as two distinct events in the safety report. Included in this table are TEAEs of any grade that occurred in at least 10% of the patient population. Events are listed in decreasing order of frequency by any grade.

- A total of 11 grade ≥3 immune-related adverse events occurred in eight patients (13.6%):
 - Pneumonitis (n=3; 5.1%), and polyarthritis, aseptic meningitis, colitis, confusional state, diarrhea, decrease lymphocyte count, hypophysitis, and neck pain (each n=1; 1.7%).
- Three patients were previously reported to have TEAEs resulting in death; the deaths were considered unrelated to study treatment.[5] There are no new TEAEs resulting in death in this 12-month follow-up analysis.

Conclusions

- This analysis demonstrates substantial antitumor activity and increasing duration of response with cemiplimab 3 mg/kg Q2W in patients with metastatic CSCC.
 - Median duration of response has not been reached. Among responding patients, estimated 12-month duration of response was 88.9%.
- Cemiplimab 3 mg/kg Q2W had an acceptable safety profile in patients with metastatic CSCC. There were no new safety signals compared with the primary analysis.[5]
- Combined with the primary analysis of the patients with locally advanced CSCC (Group 2) from the Phase 2 study (see poster #6015), these results indicate that advanced CSCC tumors, whether metastatic or locally advanced, derive durable clinical benefit from cemiplimab.

References

1. Rogers HW et al. *JAMA Dermatol*. 2015;151:1081–1086.
2. Lomas A et al. *Br J Dermatol*. 2012;166:1069–1080.
3. Burova E et al. *Mol Cancer Ther*. 2017;16:861–870.
4. Regeneron. Libtayo® (cemiplimab-rwlc) prescribing information. Available at https://www.regeneron.com/sites/default/files/Libtayo_FPI.pdf [accessed March 20, 2019].
5. Migden MR and Rischin D et al. *N Engl J Med*. 2018;379:341–351.
6. Eisenhauer EA et al. *Eur J Cancer.* 2009;45:228–247.

Acknowledgments

The authors would like to thank the patients, their families, all other investigators, and all investigational site members involved in this study.

The study was funded by Regeneron Pharmaceuticals, Inc., and Sanofi. Medical writing support and typesetting was provided by Emmanuel Ogunnowo of Prime, Knutsford, UK, funded by Regeneron Pharmaceuticals, Inc., and Sanofi.

For any questions regarding this poster presentation, please contact Alexander.Guminski@health.nsw.gov.au.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Poster presented at the 2019 American Society of Clinical Oncology Annual Meeting, June 1–4, Chicago, Illinois.